OPTICARE program (France) to optimize care among vulnerable PLWH in virological failure

Seang S.[1,2], Charbonnier E.[1], Ktorza N.[1], Blanc C. [1], Tubiana R.[1,2], Valantin MA.[1,2] Schneider L.[1,2], Dudoit Y.[1], Palich R.[1,2], Sellem. B[1], Faycal A.[1], Soulie C.[2,3], Marcelin A.-G.[2,3], Peytavin G.[4], Balde A.[2], Assoumou L.[2], Katlama C.[1,2] **Contact: sophie.seang@aphp.fr**

[1] AP-HP, Infectious Diseases department, Pitié-Salpêtrière hospital, Paris, France

[2] Sorbonne University, Infectious Diseases department, Pitié-Salpêtrière hospital, AP-HP, Pierre Louis Epidemiology and Public Health institute (iPLESP), INSERM 1136, Paris, France

[3] Sorbonne University, Virology department, Pitié-Salpêtrière hospital, AP-HP, Pierre Louis Epidemiology and Public Health institute (iPLESP), INSERM 1136, Paris, France

[4] AP-HP, Pharmacology Department, Bichat-Claude Bernard Hospital, IAME INSERM UMR-S 1137, Paris, France

AS-EACS-20221-00304

Purpose: Despite large and free access to HIV care in France, about 10% of PLWH still remain virally uncontrolled particularly among PLWH in vulnerable situations.

We aimed to assess whether an intervention program could reengage these patients to medical care and achieve viral suppression.

Methods: OPTICARE s a single center interventional study* which includes :

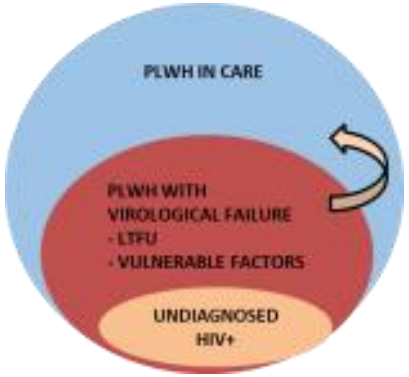
PLWH IN CARE
PLWH WITH
VIROLOGICAL FAILURE
- LTFU
- VULNERABLE FACTORS
UNDIAGNOSED
HIV+

(1) **a proactive screening of patients** in virological failure (VF) and lost to follow up (LTFU)

(2) an individualized support with a **multidiscipli-nary approach** (HIV physician, educational nurse, psychologist, social worker, cultural mediator) and **D0,week(W)4, W12, W24, W36, W48 visits.**

OPTICARE enrolled (January 2019-January 2021) **PLWH with VF** (Table 1) **AND** (1) **LTFU or** (2) with **AIDS defining illnesses or** (3) **vulnerable factors.** Primary end point was the proportion of patients retained in care and with plasma viral load (pVL) <50 cp/ml at W48**. We report W24 results.**

**ClinicalTrials.govNCT03799276*

Table 1: OPTICARE program inclusion/exclusion criteria

*LTFU : 2 consecutive missing visits, § alcohol consumers : > 40g/day (women) or > 60g/day (men)

Inclusion criteria	Exclusion criteria
1 - **HIV -1 patients in virological failure** (2 consecutive pVL >50cp/mL in the year previous the program enrollment) **AND one of the following circumstances:** **A- LTFU** * situation defined as no visit: In the 12 past months when CD4 >250/mm3 or In the 6 past months when CD4 <250/mm3 or In the 3 past months following new HIV diagnosis **B- AIDS defining illness** (< 3months) in a context of antiretroviral interruption **C- Vulnerability risk factor among** (at least one): - Social frailty (lack of health insurance, unstable accommodation) - Migrants (< 6 months arrival in France) - Frailty situation (3 months postpartum period, history of incarceration, psychiatric disease, active intravenous drug users, alcohol consumers§) **2- HIV-2 patients <200/mm3 CD4 cells,** regardless level of pVL	1-study refusal 2- HIV-1 pVL<50 cp/mL over one year

OPTICARE intervention

n=219 eligible patients

n=70 unreachabled considered as LTFU
n=28 refusal
n= 11 missed D0 OPTICARE

n=110 patients enrolled in the OPTICARE program

Graph 1: Opticare study progression over 24 weeks follow up (n=110)
* Optionnal visits (On-site or virtual)

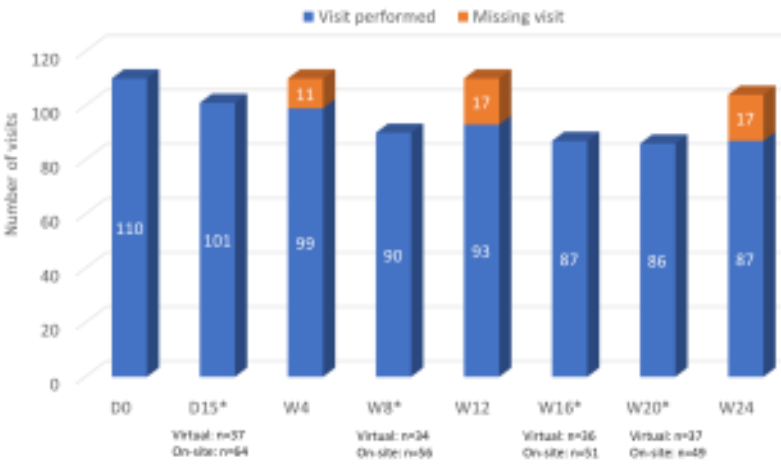
Visit performed
Missing visit
Number of visits
120
100
80
60
40
20
0
110
101
99
11
90
93
17
87
86
87
17
D0
D15*
Virtual: n=37
On-site: n=64
W4
W8*
Virtual: n=34
On-site: n=56
W12
W16*
Virtual: n=36
On-site: n=51
W20*
Virtual: n=37
On-site: n=49
W24

June 2021: **Adherence rate to the program: 89%** (10LTFU, 1 death)
Main ART strategies included 2NRTIs with INSTIS (38%) or PIs (36%) or NNRTIs (38%)
At W24, 79 (81%) had pVL <200 cp/ml ; 59 (60%) <50cp/ml

Conclusion:
The OPTICARE program showed high acceptability rate, retention in care and adherence to ART.
This work has been supported by VIIV

Table 2: Baseline biological and clinical caracteristics of OPTICARE patients

	N=110
HIV-1 (n,%)	108 (98)
Women (n,%)	60 (54)
Sub Saharian Africa (n,%)	**74 (67)**
Age (years, IQR)	42 (34-52)
Vulnerabilities (n,%)	
- Unstable accomodation	**79 (72)**
- Irregular situation	**41 (37)**
- Absence of health insurance	**25 (23)**
- ≥ 2 vulnerabilities	**65 (59)**
Psychological disorders (n,%)	**95 (86)**
Duration from HIV diagnosis (years, IQR)	**11 (5-20)**
ART duration (years, IQR)	8 (2-17)
ART interruption at baseline (n,%)	**67 (61)**
Delay of ART interruption (months, IQR)	14 (7-33)
Baseline pVL (log/mL)	**3.3 (2.3-4.7)**
Baseline CD4 (/mm3)	**190 (97-384)**
Prior AIDS (n,%)	41(37)
Genotypic HIV mutation *(n,%)	46 (42)
- NRTIs (n,%)	38 (34)
- NNRTIs (n,%)	36 (33)
- PIs (n,%)	21 (19)
- INSTIs (n,%)	19 (17)

*http://www.hivfrenchresistance.org/ 2020